Sex-Specific White Matter Microarchitectural Alterations in Preadolescent Youth With Anxiety Disorders

Nakul Aggarwal[1], Do Tromp[1], Daniel Pine[2], Lisa Williams[1], Ned Kalin[1]

[1]Department of Psychiatry, University of Wisconsin-Madison; [2]National Institute of Mental Health

Introduction

- Anxiety disorders (ADs) emerge in childhood, and, with the onset of puberty, females are twice as likely as males to have ADs.[1]
- Considerable research has implicated alterations in the white matter (WM) microstructure of prefrontal-limbic tracts, as well as other WM regions, in the pathophysiology of ADs.[2,3,4]
- These findings are of interest because WM consists primarily of myelinated axons, and myelin plays an important role in mediating optimal neuronal communication.
- Few studies have examined AD-related WM alterations in youth and in relation to sex.
- To further examine associations between childhood ADs, WM microstructural integrity, and sex, we analyzed diffusion tensor imaging (DTI) data from 295 preadolescent youth with and without ADs.

Methods

- Across 3 studies, preadolescent youth with ADs (social, generalized, and/or separation AD) and healthy controls were enrolled between the ages of 8-12 at 2 study sites (UW-Madison and NIMH) and completed a DTI scan on a 3T-scanner.
- The final sample (n=295; 201 F, 94 M) included 163 children with ADs and 132 healthy controls.
- Using a mega-analytic approach, tractography-based and voxelwise analyses examined the main effect of ADs, as well as the AD-by-sex interaction, in relation to 4 WM metrics (fractional anisotropy [FA], radial diffusivity [RD], mean diffusivity [MD], and axial diffusivity [XD]) in 7 bilateral tracts of interest and in WM across the whole brain.

Results

- Tract-based and voxelwise results demonstrated widespread AD-related alterations in DTI metrics across multiple WM regions.
- Critically, AD-related effects demonstrated a significant interaction with sex, such that FA reductions and RD increases were observed exclusively in boys and not in girls, indicating anxiety-related WM alterations may be more robust in males than in females.
- Preadolescent boys with ADs exhibited exhibited decreased FA and elevated RD relative to healthy control boys (P<0.05, corrected) across various WM regions across the brain, in association (UF, EC, IFO, SLF, ST, ILF), commissural (CC), projection (CR, IC), and brainstem (CST, CP, ML) tracts; no group differences were seen in girls.

Discussion

- This study constitutes the largest cross-sectional DTI study of childhood anxiety to date.
- Results show that childhood ADs are associated with broadly distributed alterations in WM microstructure and, importantly, this relationship appears more robust or prominent in boys.
- The findings – particularly the combination of reduced FA and increased RD – point to the possibility that altered myelination processes are associated with childhood ADs.
- While sex-related hormone levels did not account for the sexually dimorphic effect, *in vitro* evidence suggests differential sensitivity to stress in male vs. female OPCs.[5,6]
- Future work should investigate the extent to which these findings may play a causal role that would further support targeting WM microstructure, especially in boys with ADs.

Acknowledgements

We thank the participants and families, as well as the staff of the HealthEmotions Research Institute at the Wisconsin Psychiatric Institute and Clinics and the Section on Developmental and Affective Neuroscience at the NIMH. This research was supported by NIH grants R21MH092581, R01MH107563, U01MH112913, T32MH018931, T32GM140935, and IRP project ZIA-MH002781, as well as UL1TR002373 from the NIH NCATS CTSA program.

Tract-Based Analyses – Sex-Specific AD-Related Reductions in WM FA – *P-values*

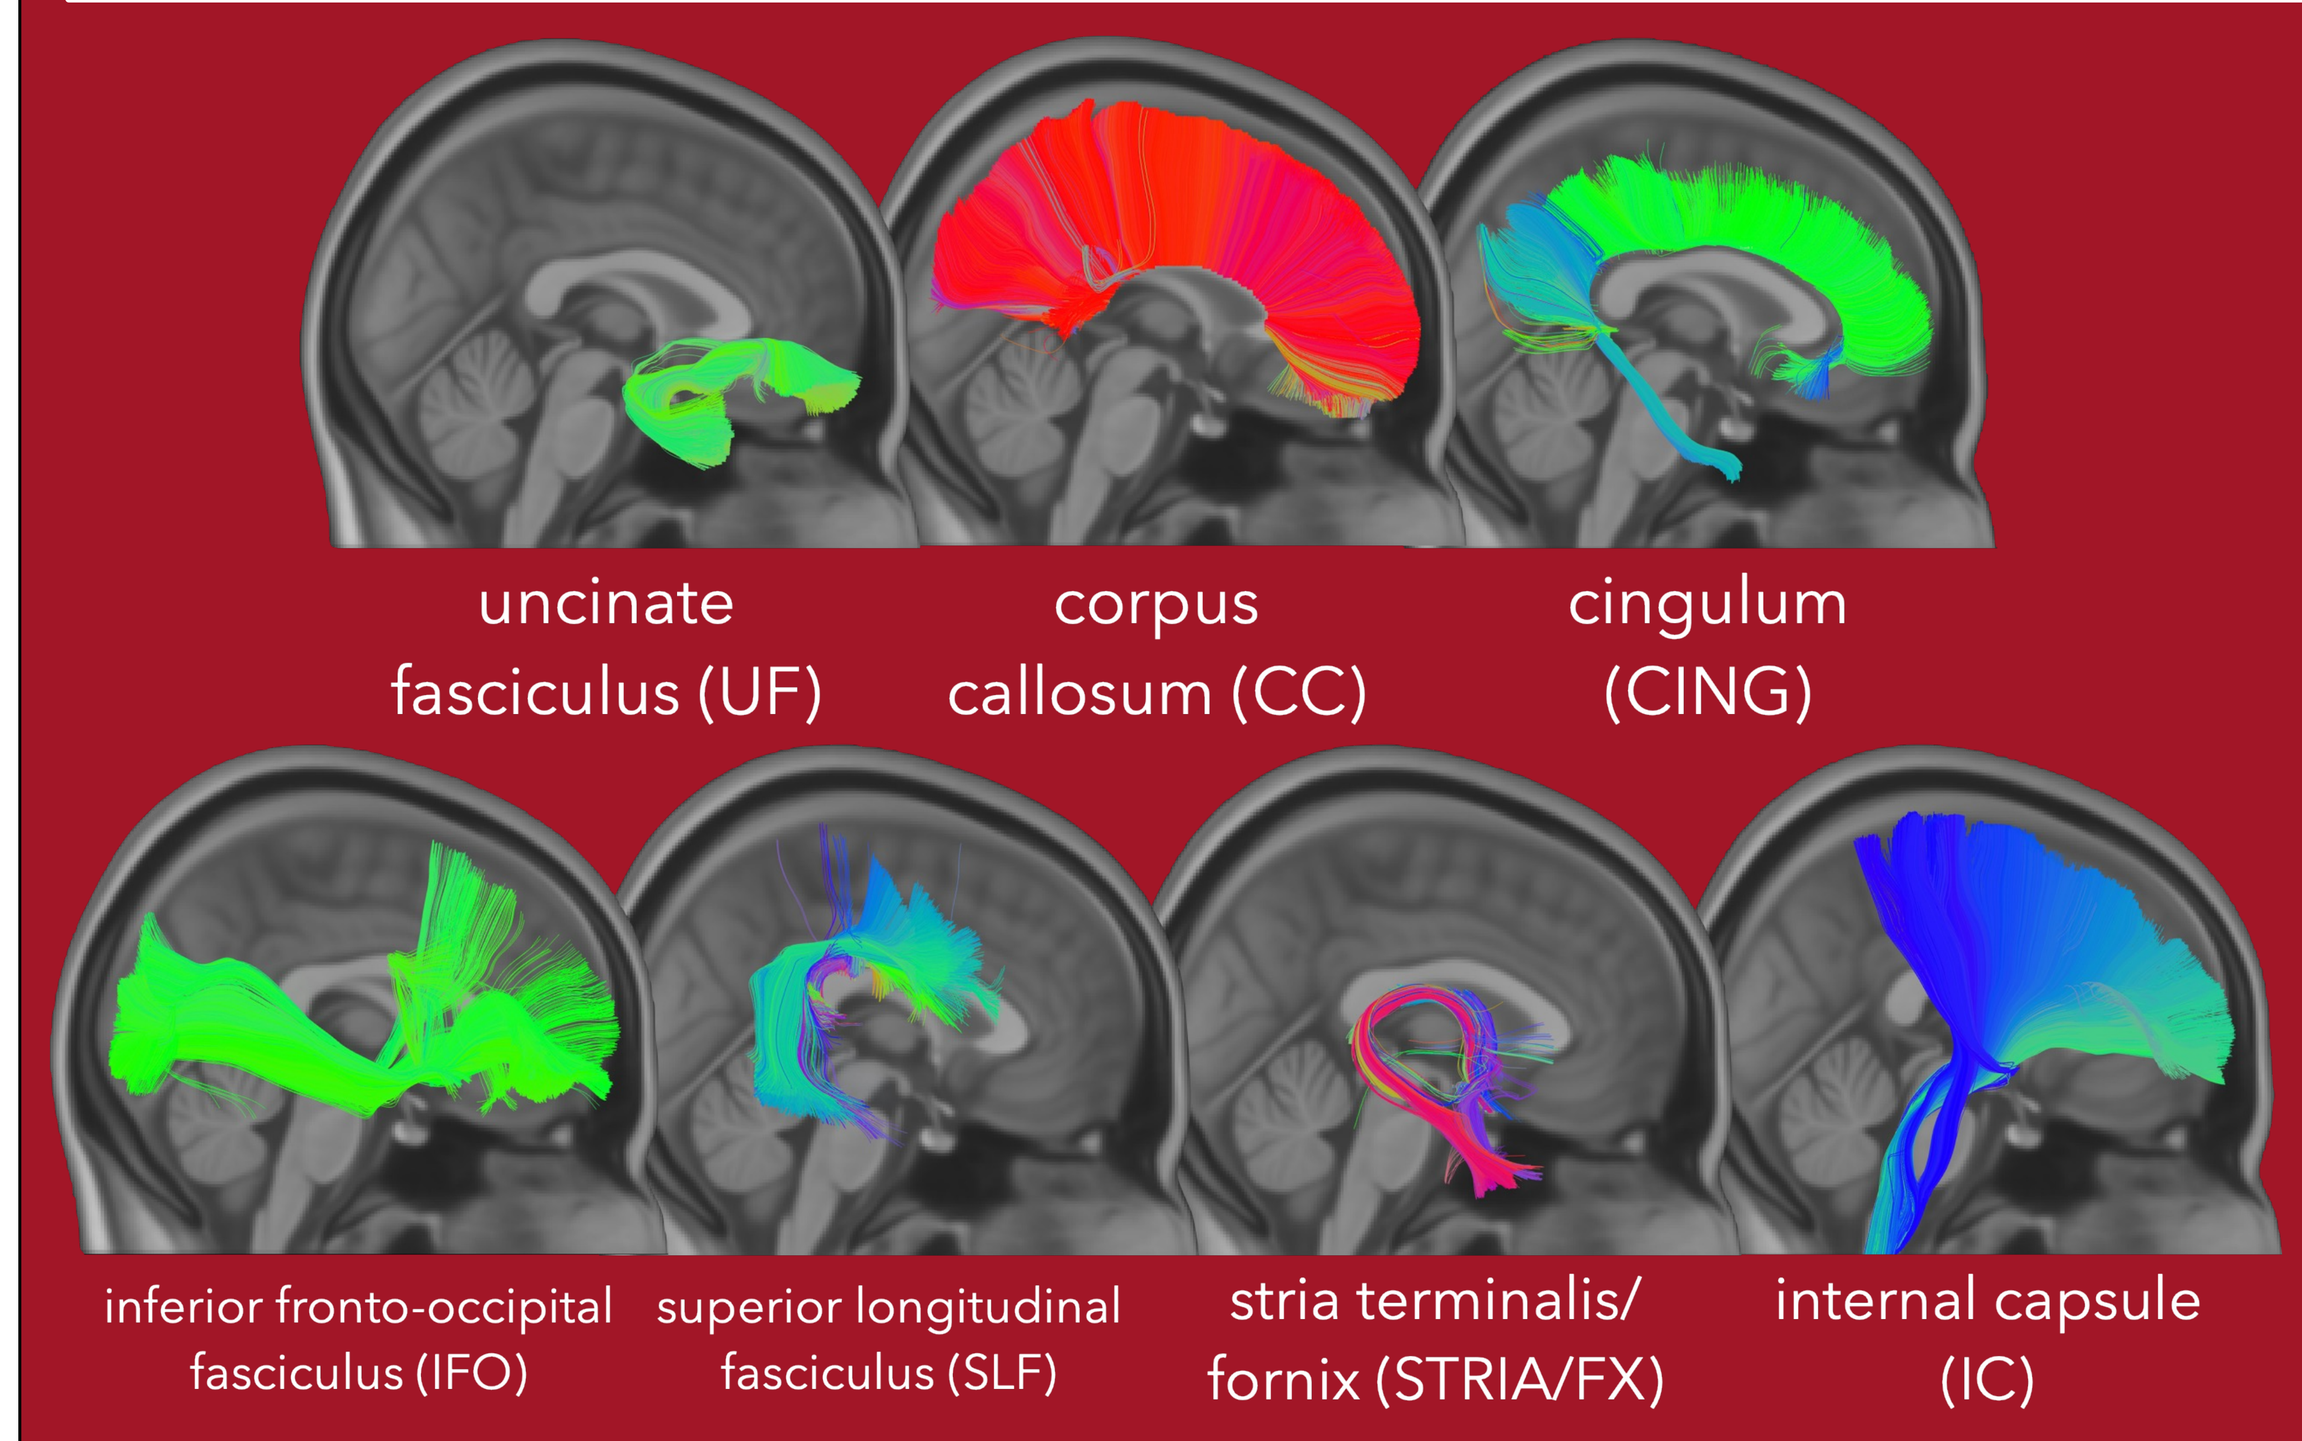
uncinate fasciculus (UF)
corpus callosum (CC)
cingulum (CING)
inferior fronto-occipital fasciculus (IFO)
superior longitudinal fasciculus (SLF)
stria terminalis/ fornix (STRIA/FX)
internal capsule (IC)

= p<0.05, Bonferroni corrected (green)
= p<0.05, uncorrected (yellow)

FA	N=295		MALES	FEMALES
Tract	**Group**	**Group x Sex**	**Group**	**Group**
UF	0.00236	0.00184	0.000411	0.931565
CC	0.01421	0.00789	0.00218	0.842
CING	0.0713	0.0697	0.0181	0.945
IFO	0.02090	0.00252	0.00104	0.515
SLF	0.1138	0.0112	0.00783	0.384
STRIA/FX	0.00639	0.02538	0.00367	0.660
IC	0.5926	0.0302	0.06617	0.178

AD children exhibit **widespread, distributed reductions** in white matter microstructure, and these alterations appear **more robust in males**.

Voxelwise Analyses – Sex-Specific AD Effects on WM FA and RD

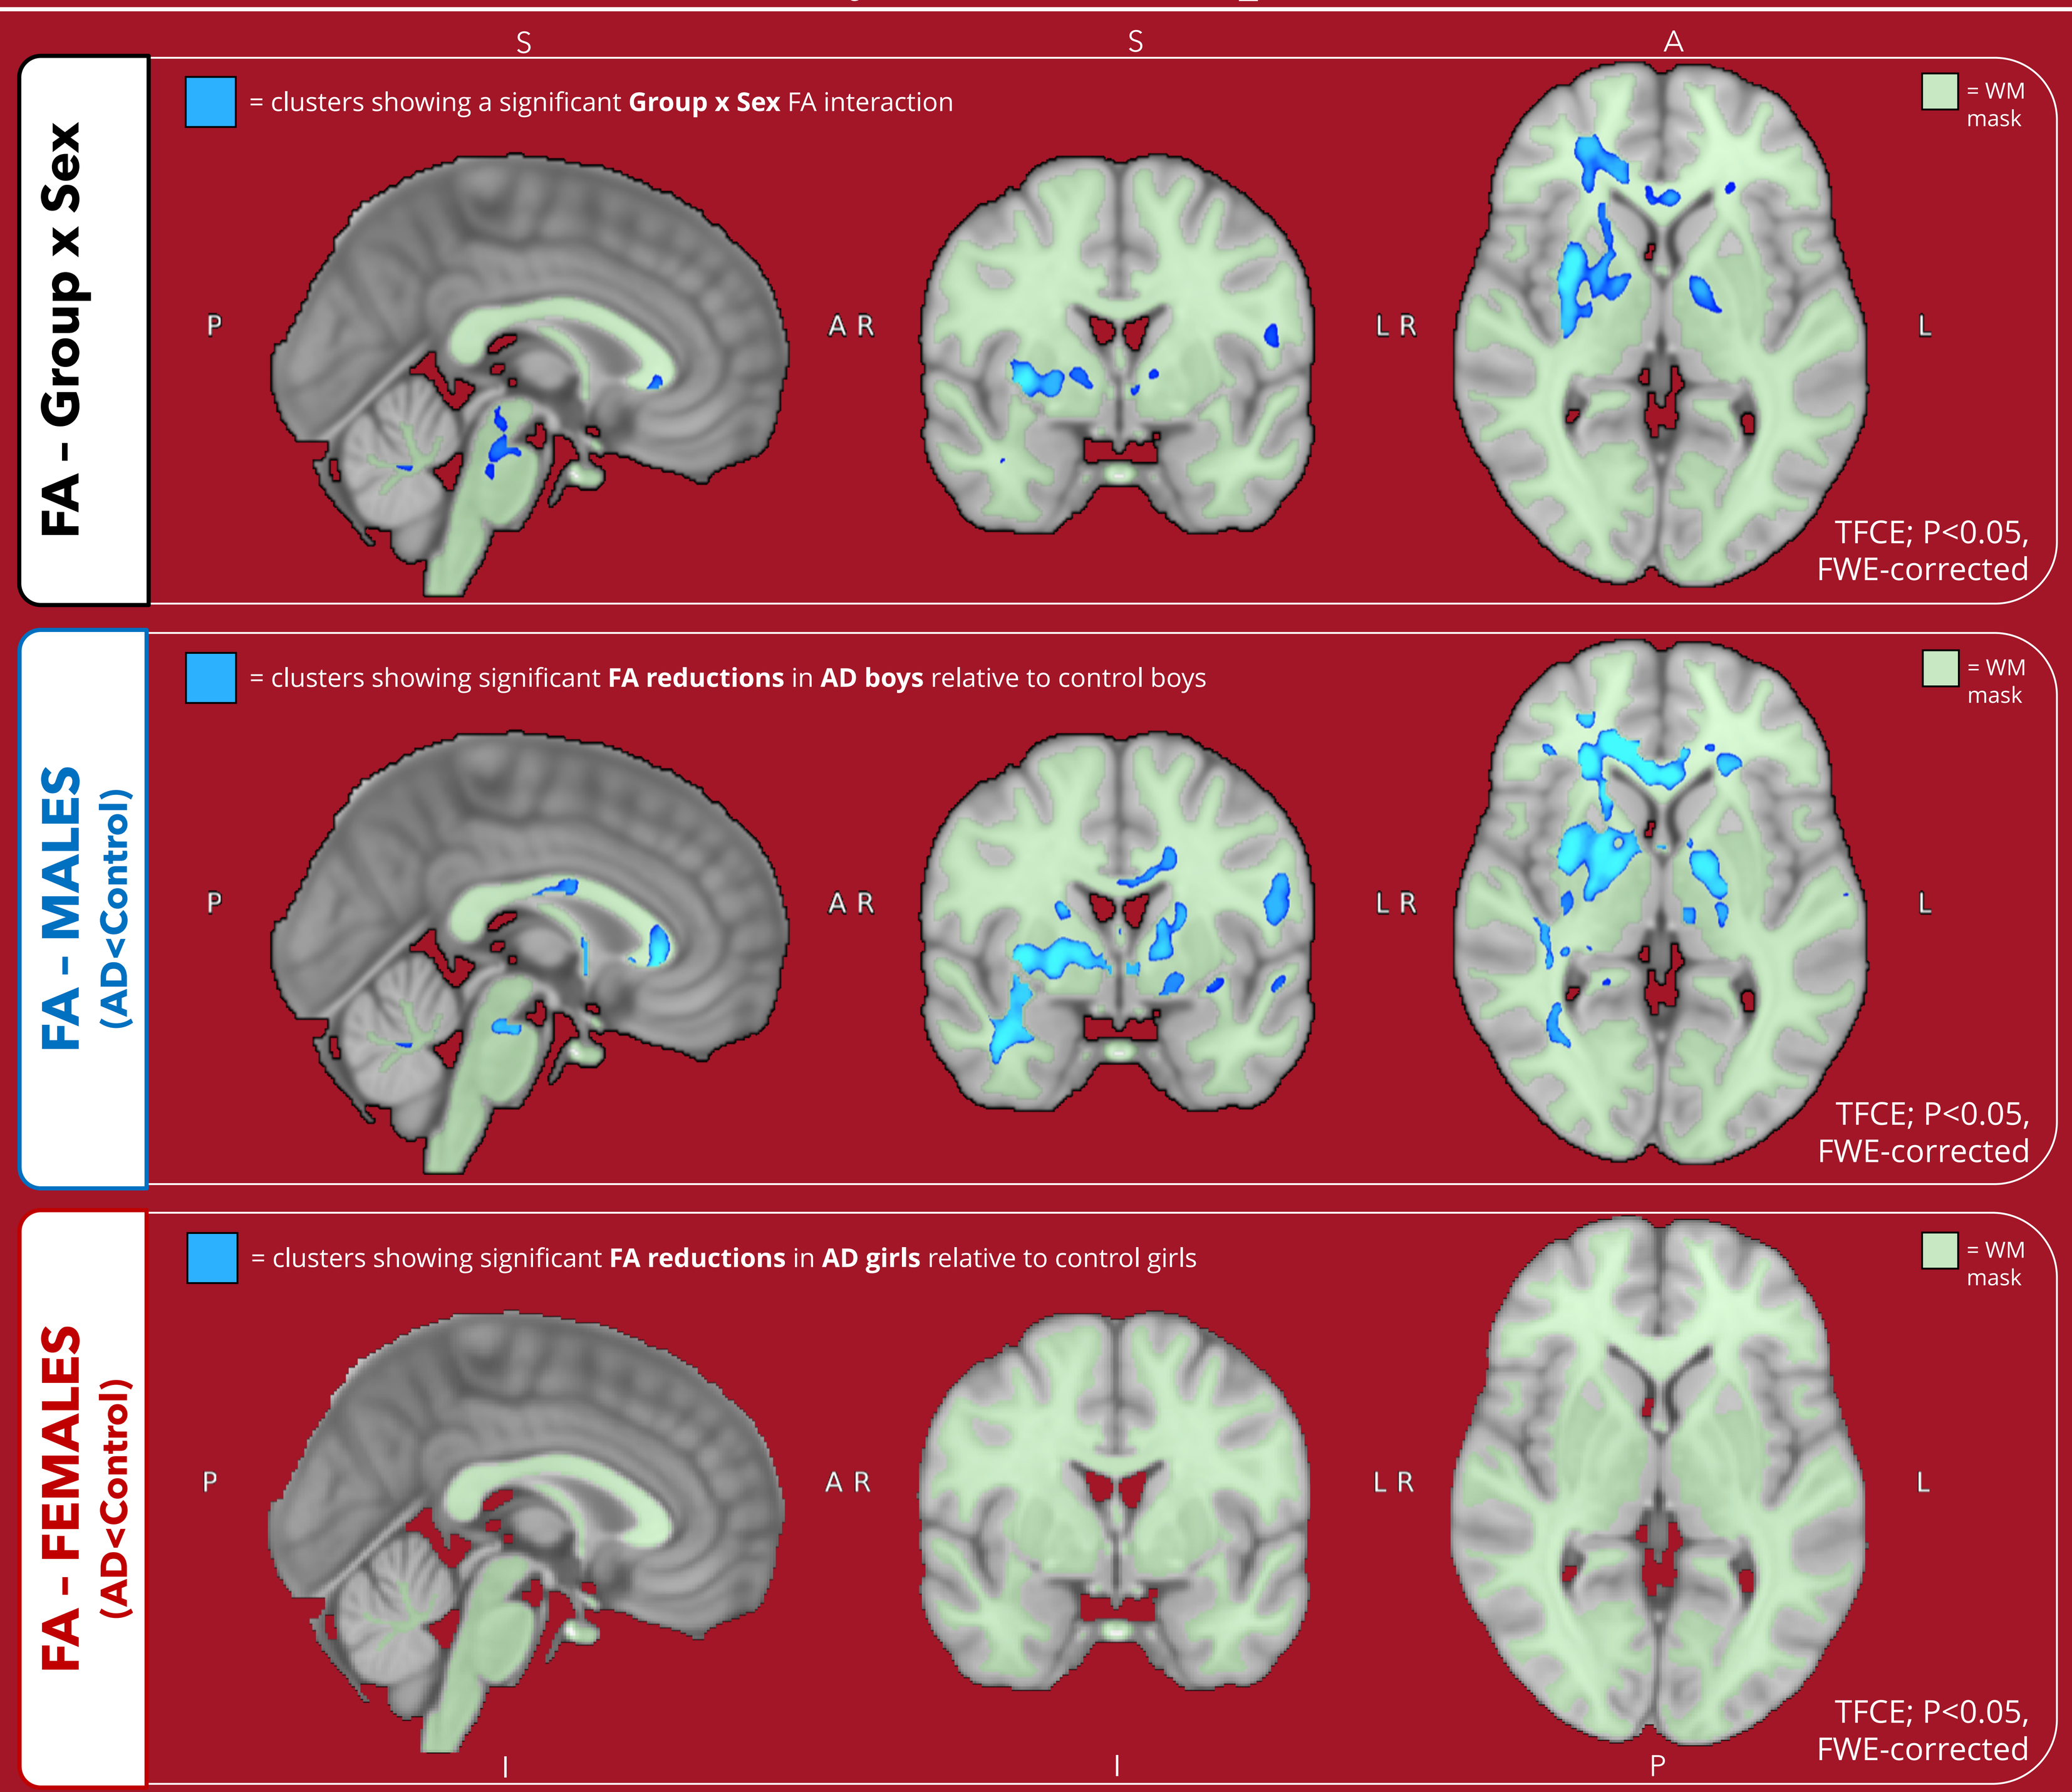
FA - Group x Sex
= clusters showing a significant **Group x Sex** FA interaction
FA - MALES (AD<Control)
= clusters showing significant **FA reductions** in **AD boys** relative to control boys
FA - FEMALES (AD<Control)
= clusters showing significant **FA reductions** in **AD girls** relative to control girls
= WM mask
TFCE; P<0.05, FWE-corrected

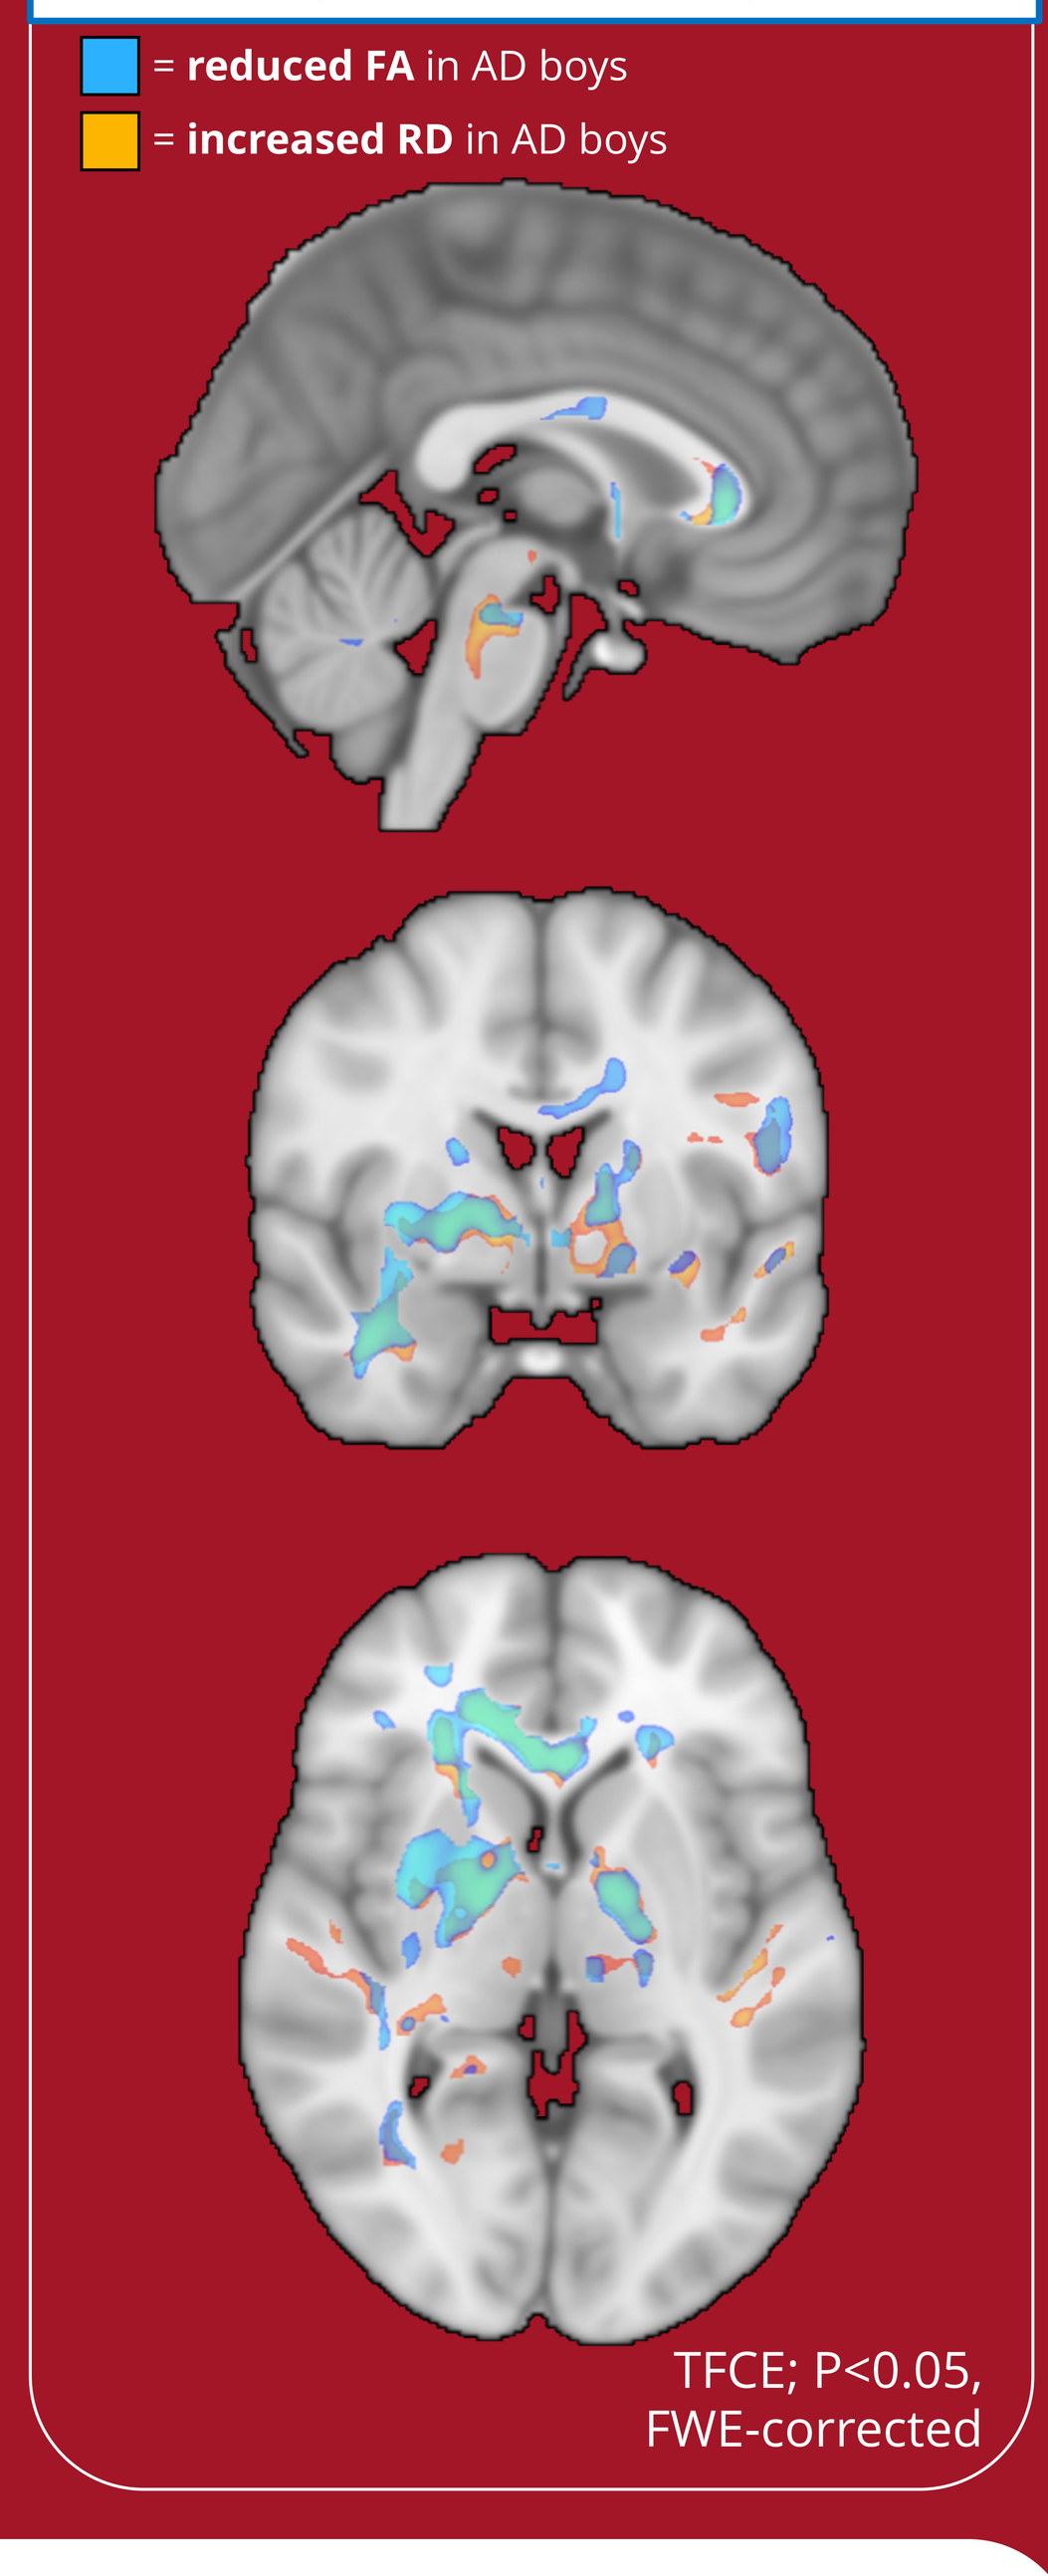
FA and RD Changes Overlap
MALES
(AD vs. Control)
= **reduced FA** in AD boys
= **increased RD** in AD boys
TFCE; P<0.05, FWE-corrected

Proposed **theoretical framework** for explaining the sexually dimorphic WM-anxiety associations

⟶ *sex-based differences in OPC biology*

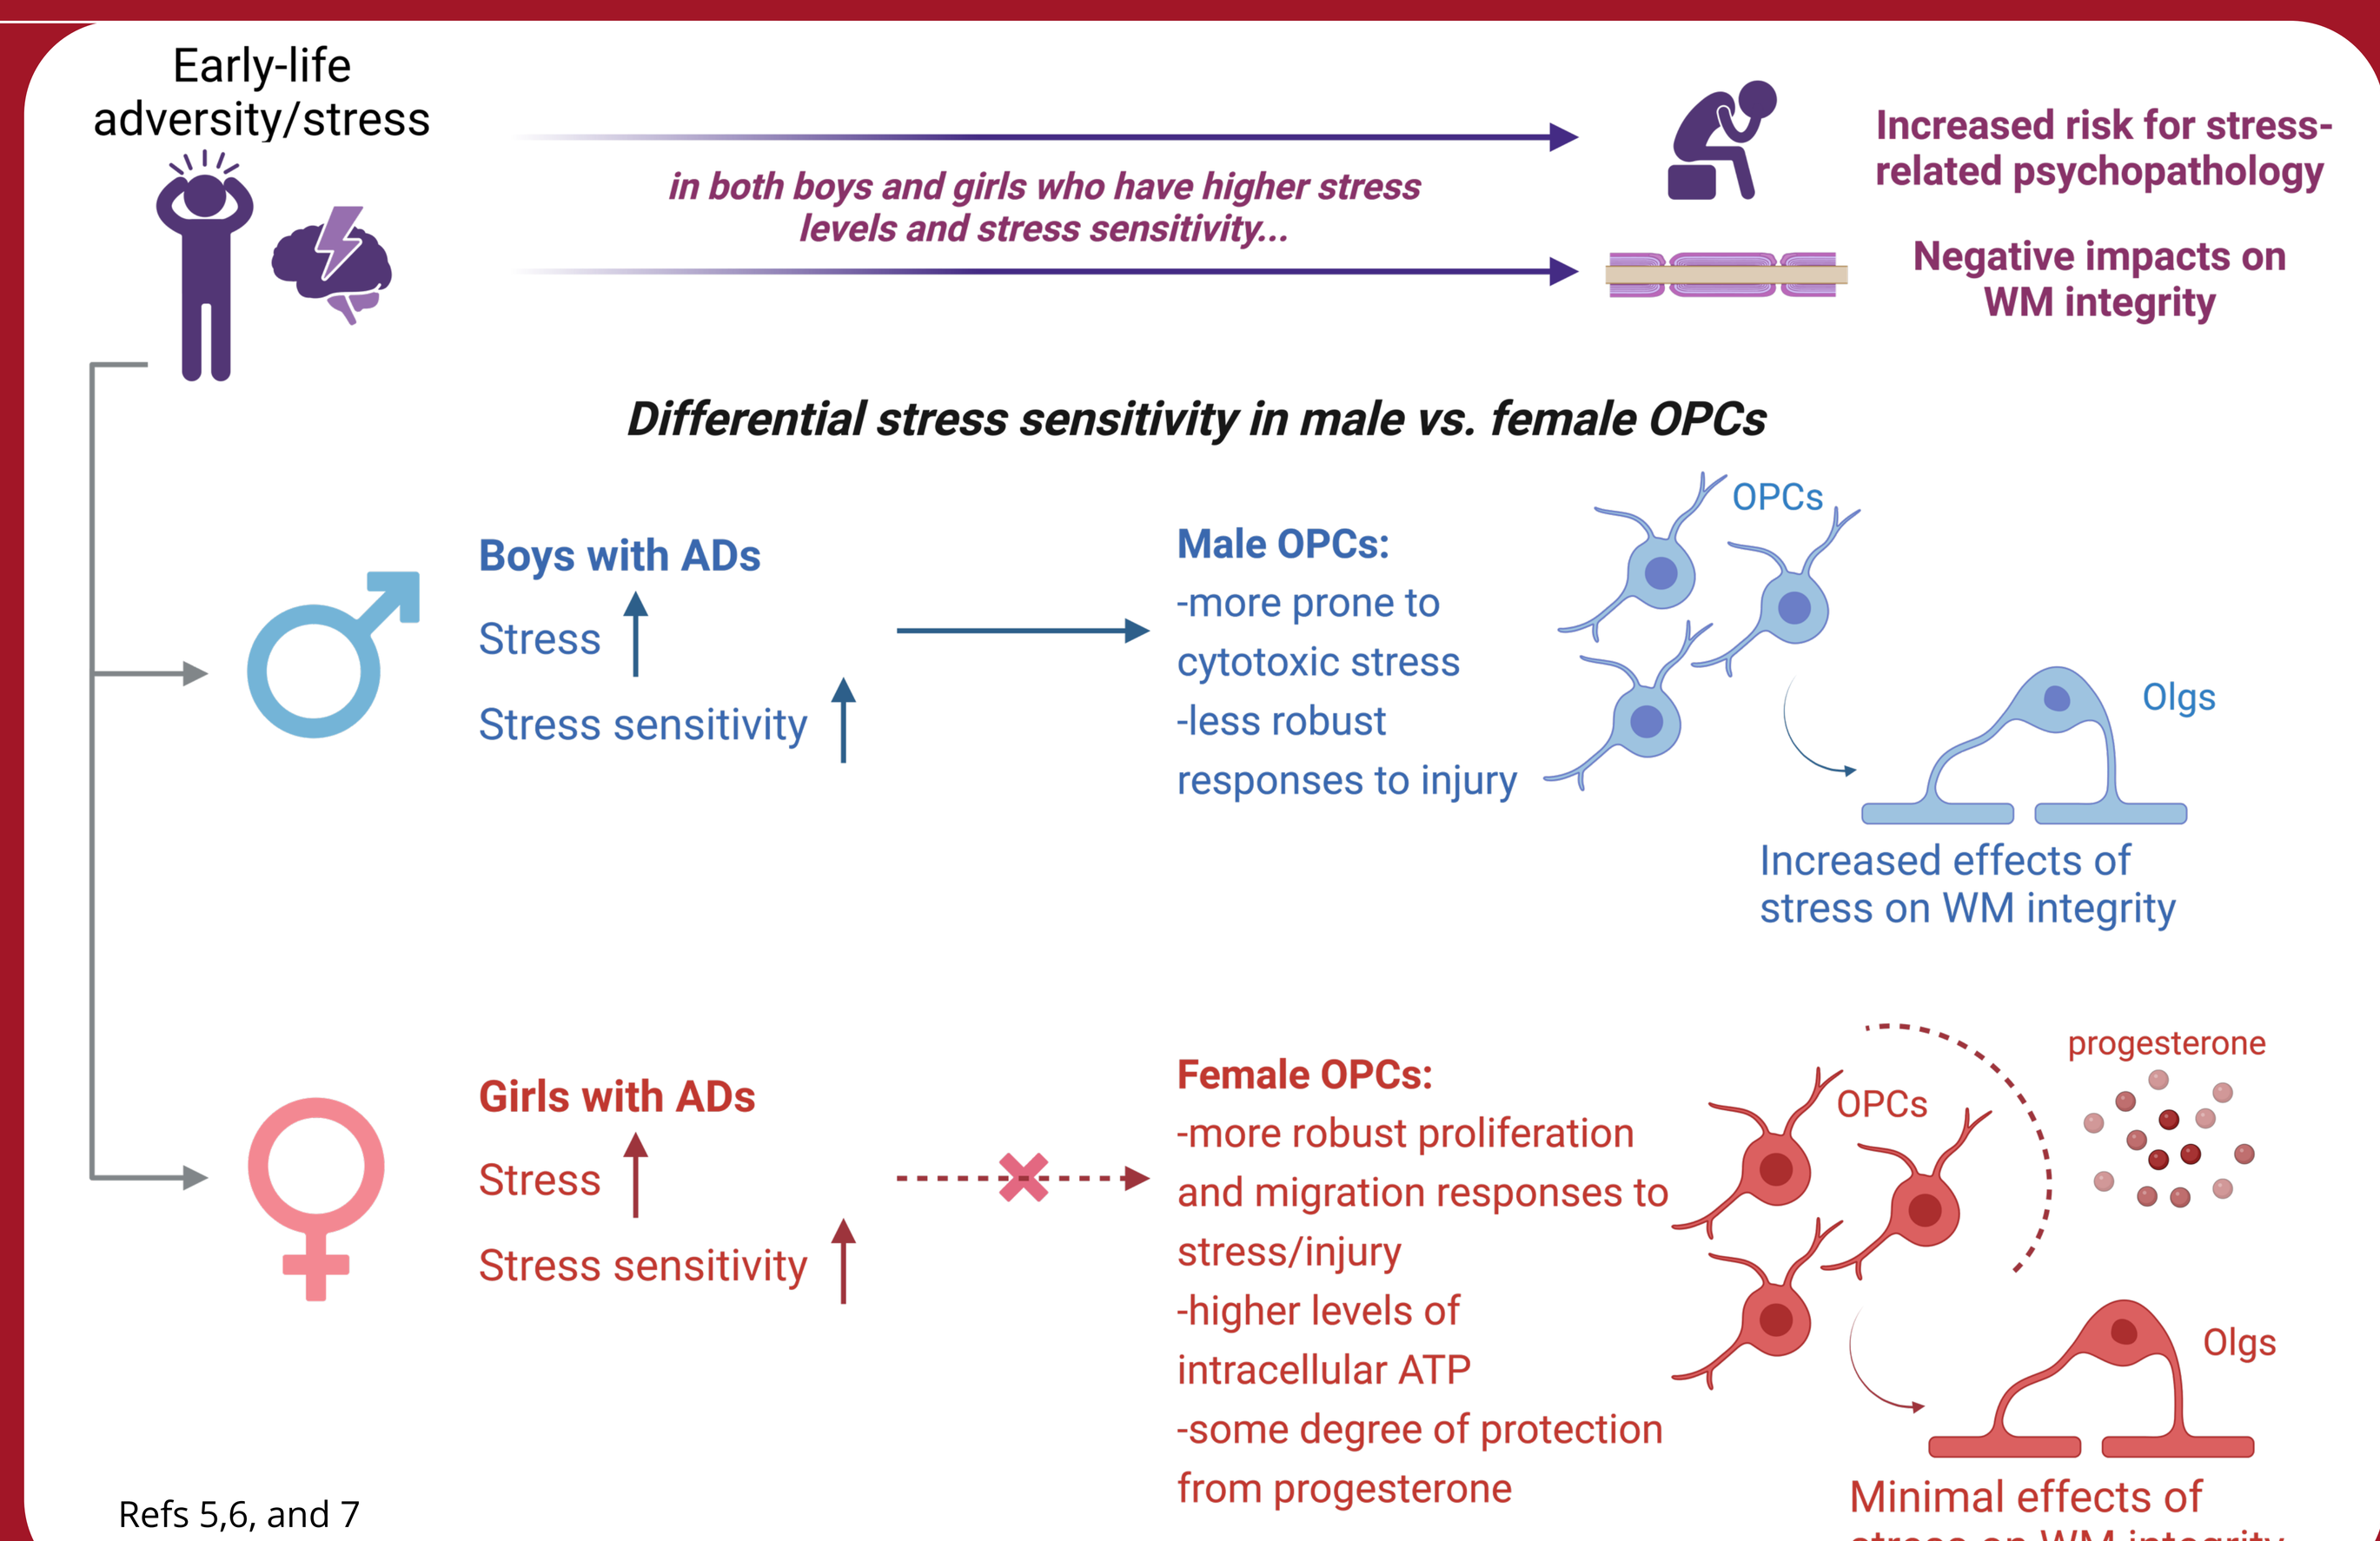
Early-life adversity/stress
in both boys and girls who have higher stress levels and stress sensitivity...
Increased risk for stress-related psychopathology
Negative impacts on WM integrity
Differential stress sensitivity in male vs. female OPCs
Boys with ADs
Stress
Stress sensitivity
Male OPCs:
-more prone to cytotoxic stress
-less robust responses to injury
OPCs
Olgs
Increased effects of stress on WM integrity
Girls with ADs
Stress
Stress sensitivity
Female OPCs:
-more robust proliferation and migration responses to stress/injury
-higher levels of intracellular ATP
-some degree of protection from progesterone
progesterone
Minimal effects of stress on WM integrity

Refs 5,6, and 7
Created with BioRender

Sample and Analysis

Demographics/Clinical Characteristics

Measure	Full Sample				Girls		Boys	
	Control Group		AD Group		Control Group	AD Group	Control Group	AD Group
	N	% Female	N	% Female	N	N	N	N
Overall sample	132	66	163	70	87	114	45	49
Demographic characteristics								
	Mean (SD)		**Mean (SD)**		**Mean (SD)**	**Mean (SD)**	**Mean (SD)**	**Mean (SD)**
Age (years)	10.53 (1.19)		10.59 (1.23)		10.58 (1.10)	10.62 (1.16)	10.43 (1.36)	10.51 (1.40)
IQ (WASI)	116.71 (13.02)		113.85 (14.57)		115.06 (13.98)	113.35 (14.95)	119.82 (10.40)	114.96 (13.78)
Tanner (Physical Development)	1.76 (0.87)		1.97 (1.00)		1.80 (0.89)	2.05 (1.02)	1.69 (0.83)	1.78 (0.92)
Clinical scales								
Parent SCARED (Anxiety)[a]	4.35 (5.11)		29.59 (11.31)		4.18 (4.41)	30.06 (10.80)	4.67 (6.30)	28.52 (12.44)
Child SCARED (Anxiety)[a,b,c]	8.92 (7.50)		29.91 (13.74)		8.63 (7.15)	32.43 (13.30)	9.49 (8.18)	24.20 (13.11)
CDI (n=193) (Depression)[a,b]	34.02 (14.47)		44.50 (17.19)		36.19 (12.63)	48.17 (14.60)	27.52 (17.73)	33.32 (19.80)
MFQ (n=93) (Depression)[a]	0.26 (0.51)		5.79 (4.51)		0.43 (0.65)	6.09 (4.90)	0.14 (0.36)	5.38 (3.94)
CPRS-R (ADHD)[a,b]	44.40 (3.77)		58.99 (11.95)		45.32 (3.88)	59.95 (12.87)	42.51 (2.73)	56.79 (9.29)

[a] Significantly higher in children with ADs compared to controls (p<0.001).
[b] Significantly higher in girls compared to boys (p<0.05).
[c] Significant group by sex interaction; higher in girls with ADs compared to boys with ADs (p<0.001).

DWI Analysis – Deterministic Tractography

1. Generate tensor images for each participant
2. Delineate WM tracts of interest based on tensor anisotropy
3. Extract FA, MD, and RD values in each region for each subject

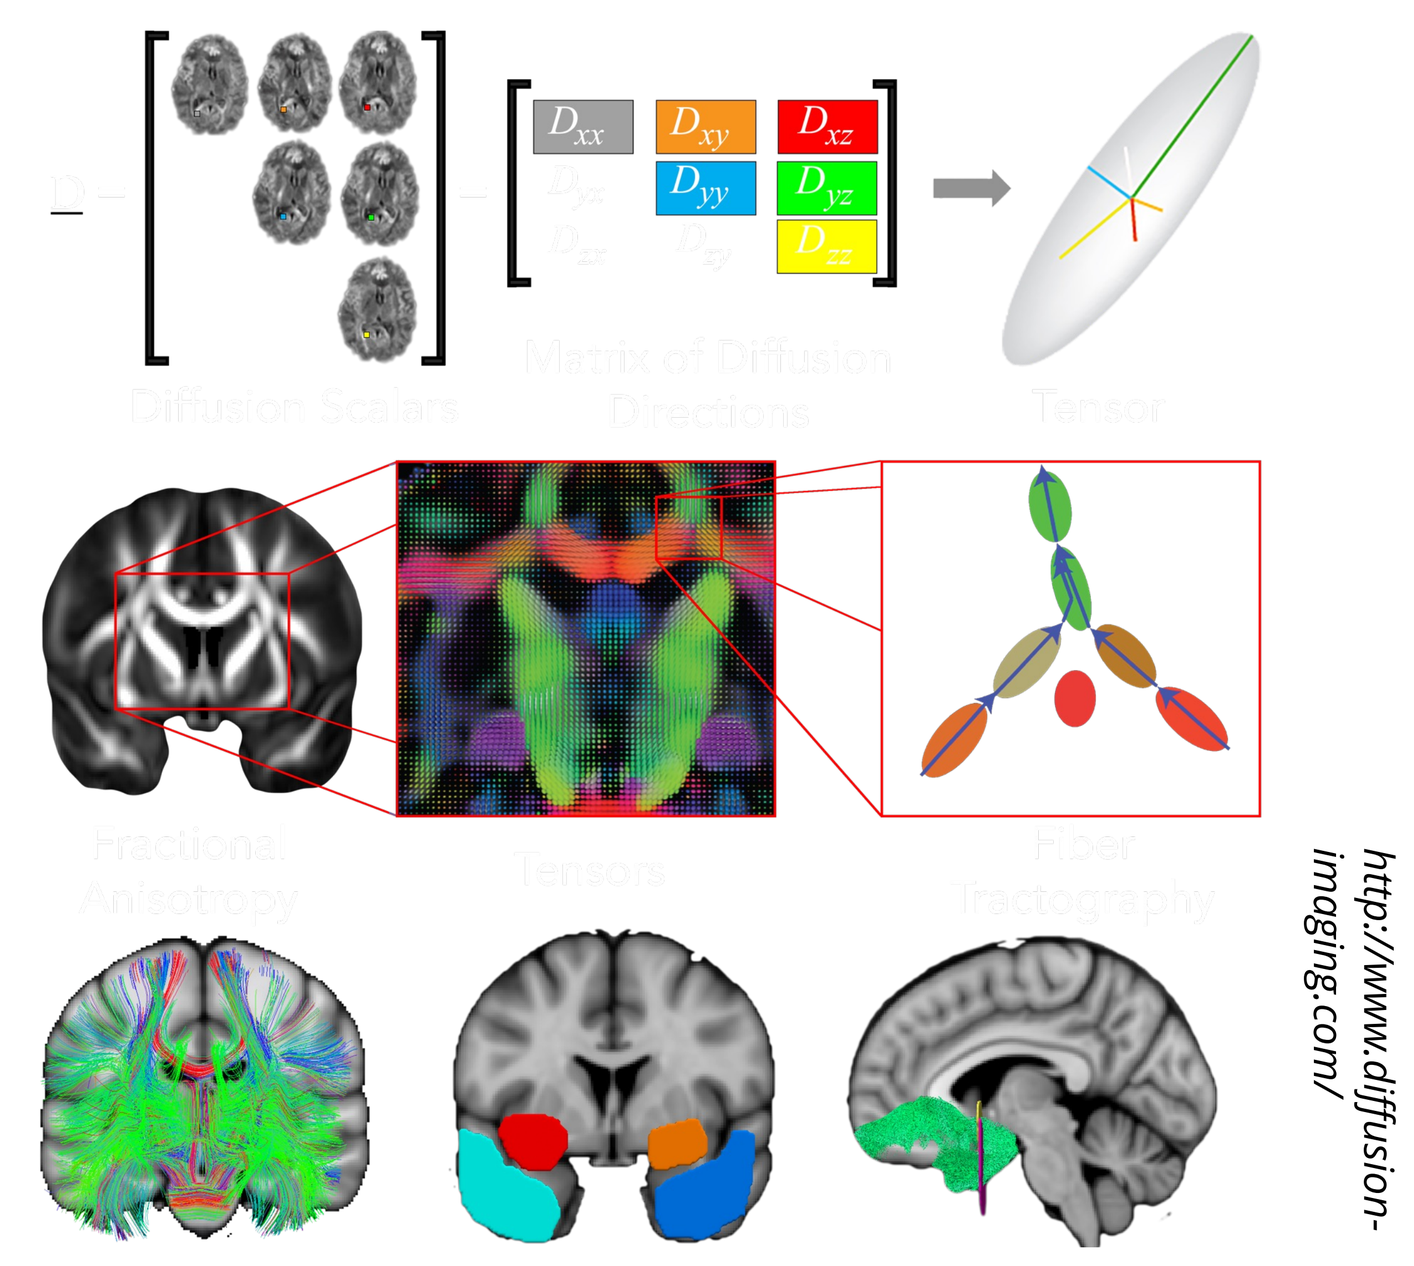
http://www.diffusion-imaging.com/

Effects of Age, Sex, and Site on Tract-Based FA Results

	FA					
	Age		Sex		Site	
Bilateral WM Tract	t	p-value	t	p-value	t	p-value
CC	5.444	<0.001	-0.474	0.636	0.364	0.716
CING	8.204	<0.001	-0.356	0.722	-0.530	0.596
IC	7.472	<0.001	-0.510	0.610	-0.095	0.924
IFO	6.509	<0.001	0.012	0.990	-2.094	0.037
SLF	5.233	<0.001	0.137	0.891	-2.315	0.021
STRIA/FX	1.210	0.227	0.055	0.956	-0.788	0.431
UF	5.021	<0.001	-0.053	0.958	-1.396	0.164

Group-by-Sex FA Interactions in 7 Bilateral WM Tracts

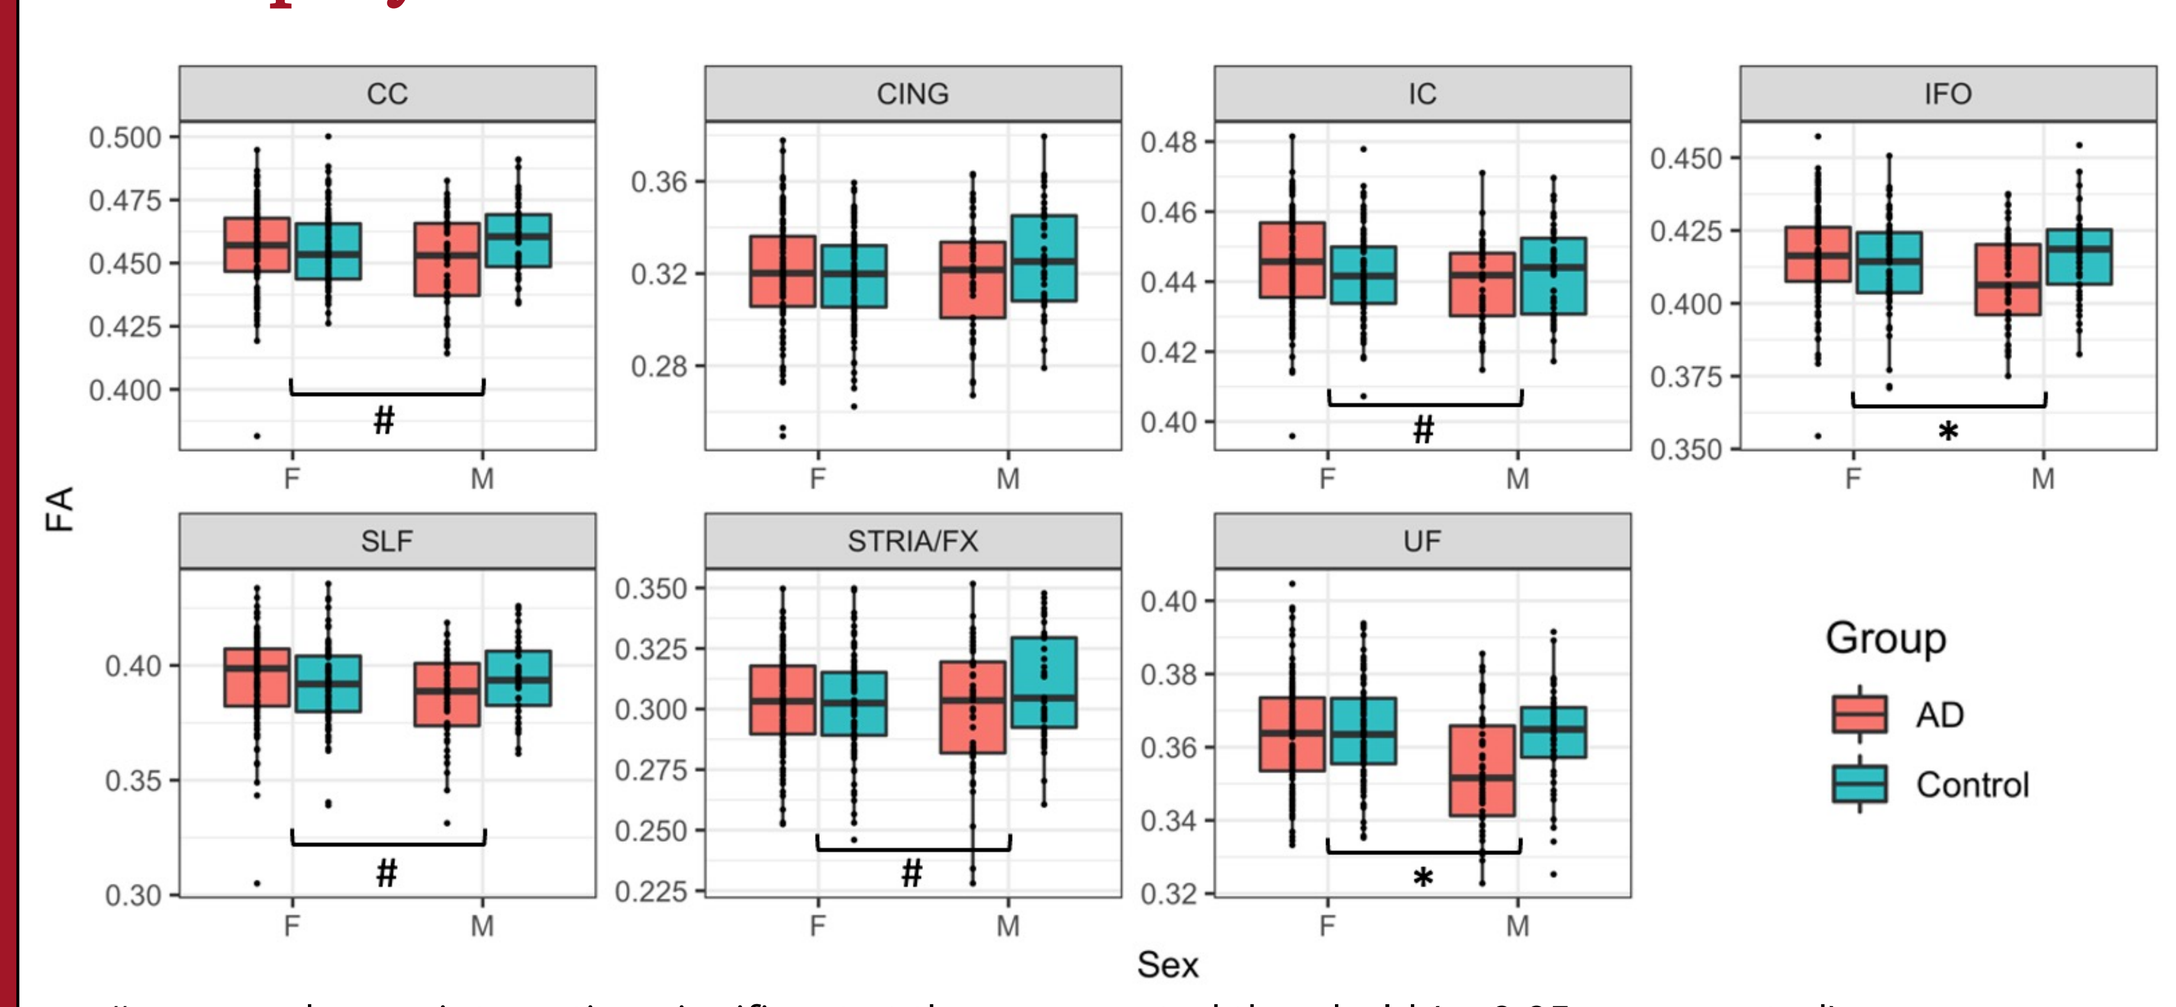
CC
CING
IC
IFO
SLF
STRIA/FX
UF
FA
Sex
F
M
Group
AD
Control

= group-by-sex interaction significant at the uncorrected threshold (p<0.05, uncorrected)
* = group-by-sex interaction significant at the Bonferroni-corrected threshold (p<0.05, corrected)

Voxelwise RD Results – Group and Group-by-Sex Effects

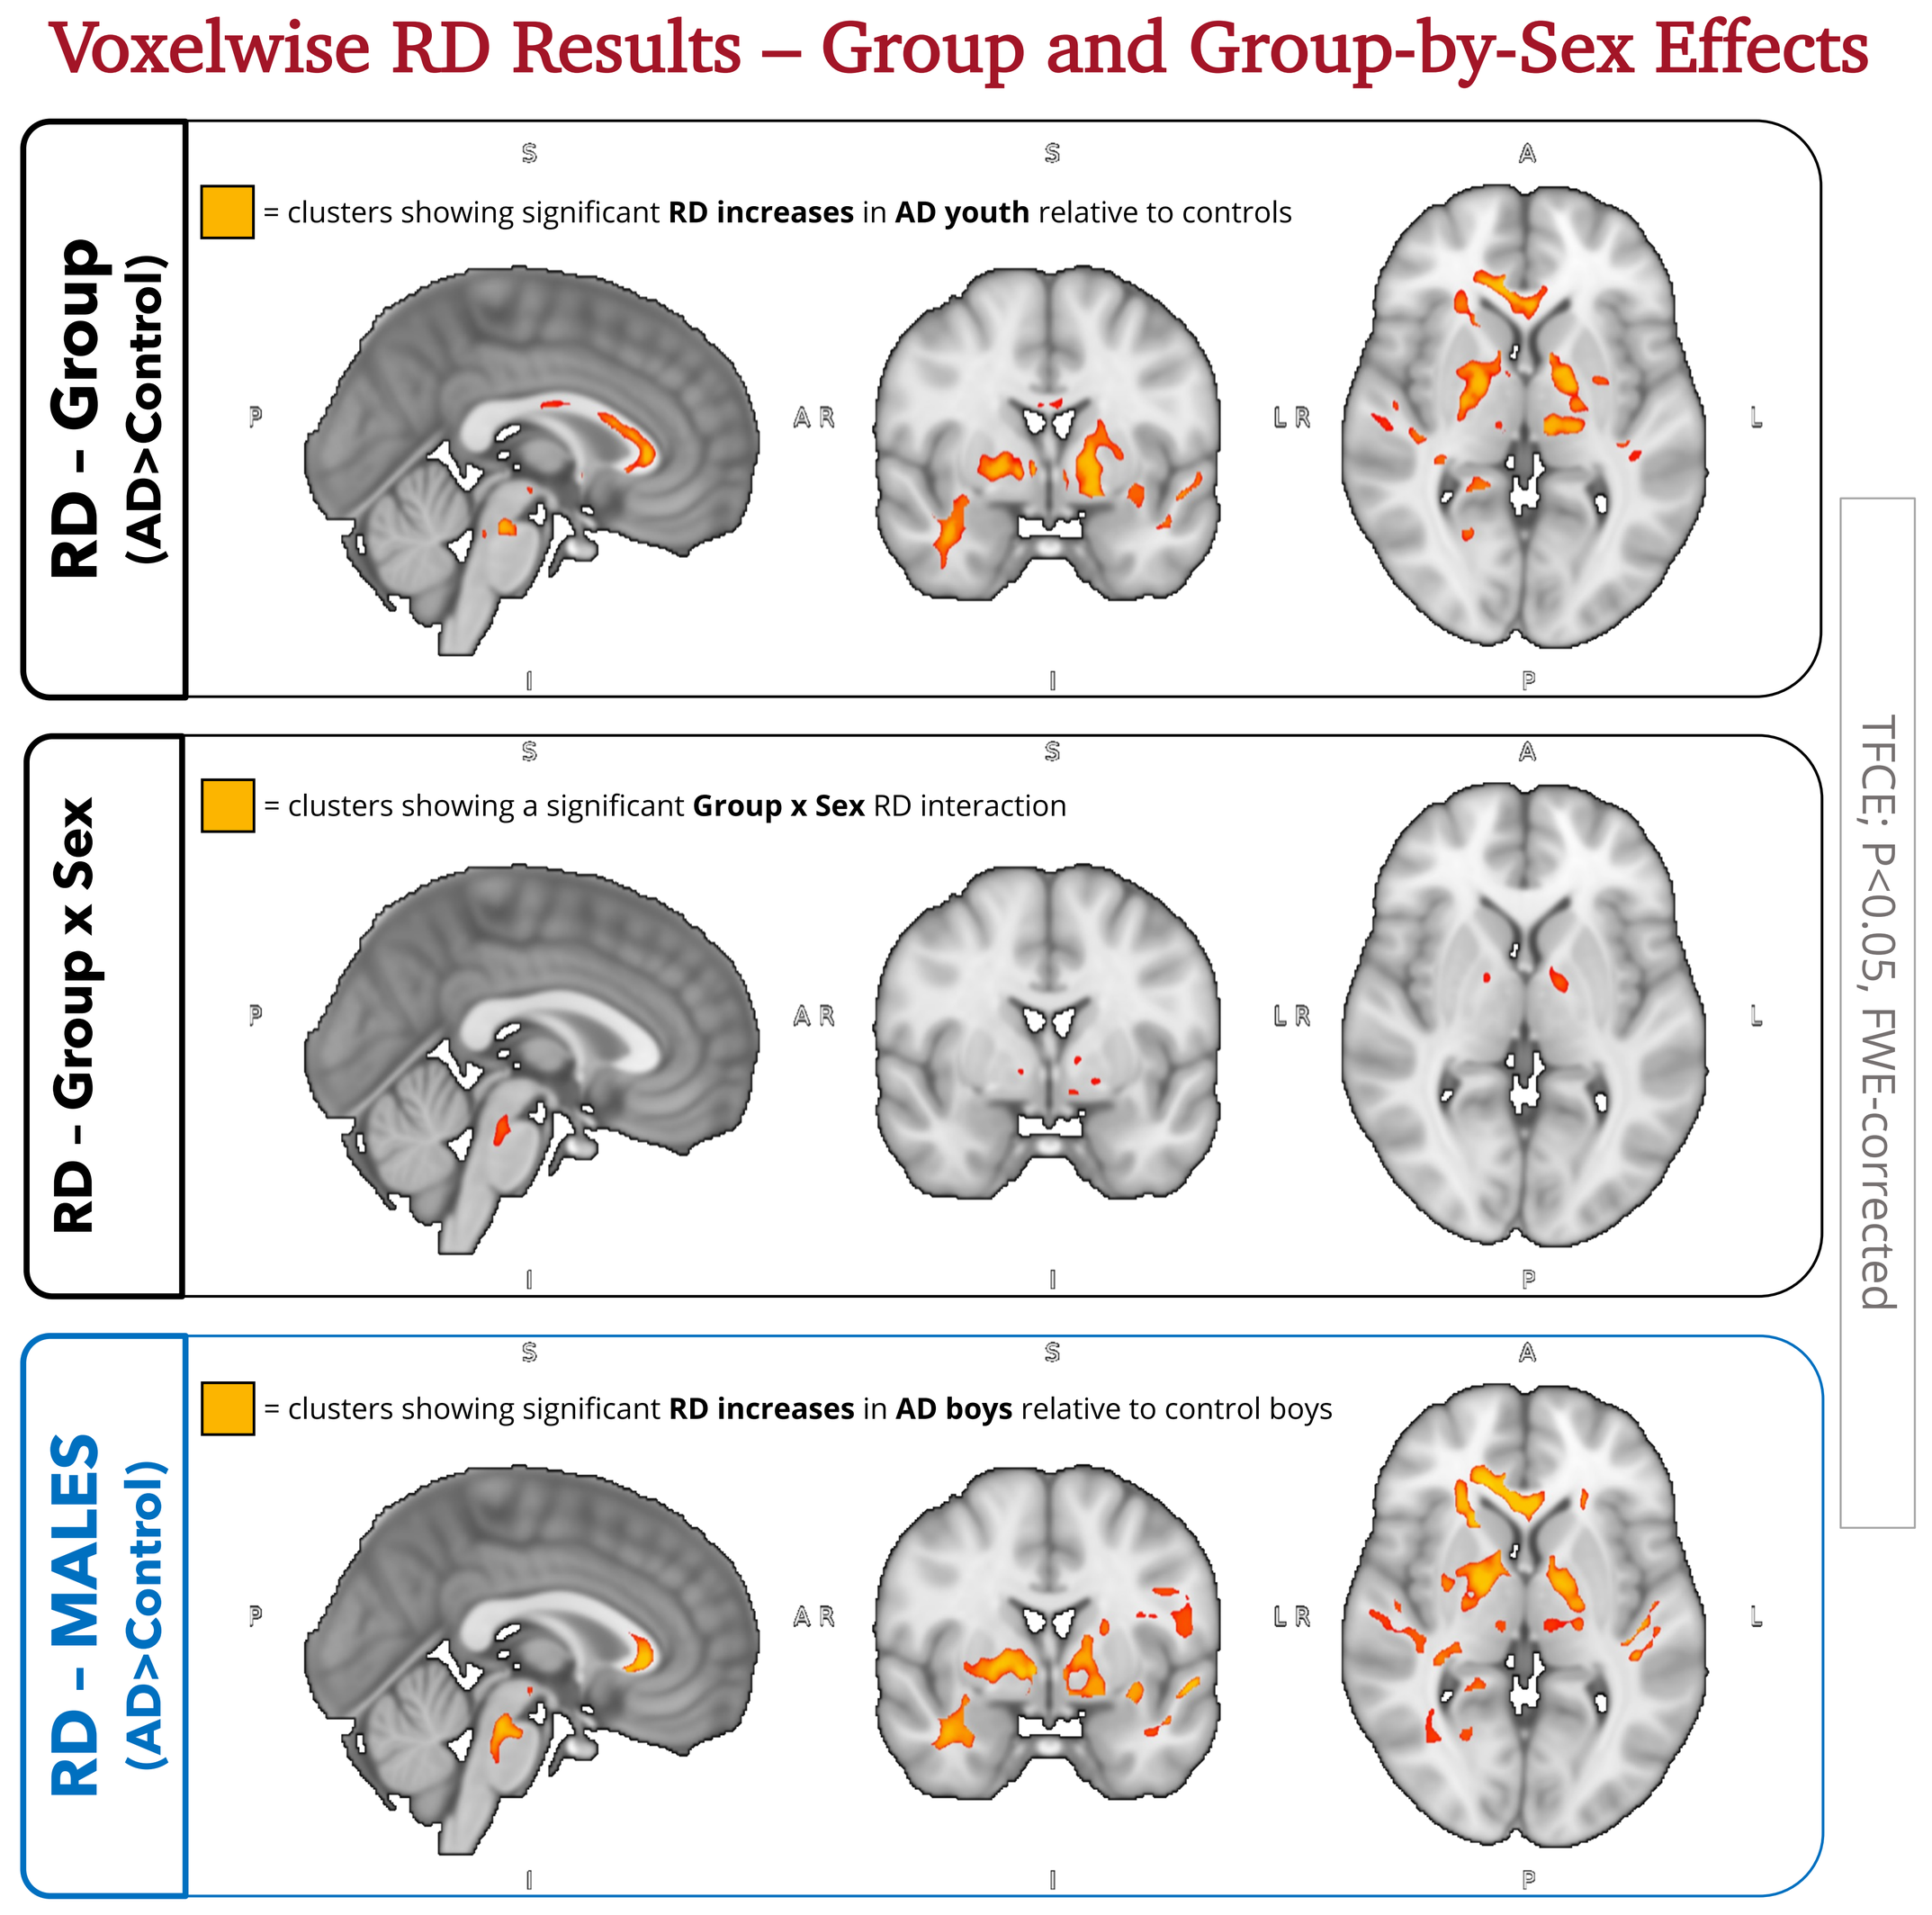
RD - Group (AD>Control)
= clusters showing significant **RD increases** in **AD youth** relative to controls
RD - Group x Sex
= clusters showing a significant **Group x Sex** RD interaction
RD - MALES (AD>Control)
= clusters showing significant **RD increases** in **AD boys** relative to control boys
TFCE; P<0.05, FWE-corrected

References
1. Kessler RC, McGonagle KA, Zhao S, Nelson CB, Hughes M, Eshleman S, et al. Lifetime and 12-Month Prevalence of DSM-III-R Psychiatric Disorders in the United States: Results From the National Comorbidity Survey. Arch Gen Psychiat. 1994;51(1):8–19.
2. Tromp DPM, Williams LE, Fox AS, Oler JA, Roseboom PH, Rogers GM, et al. Altered Uncinate Fasciculus Microstructure in Childhood Anxiety Disorders in Boys But Not Girls. American Journal of Psychiatry. 2019 Mar;176(3):208–16.
3. Liao M, Yang F, Zhang Y, He Z, Su L, Li L. White matter abnormalities in adolescents with generalized anxiety disorder: a diffusion tensor imaging study. Bmc Psychiatry. 2014;14(1):41.
4. Aggarwal N, Williams LE, Tromp DPM, Pine DS, Kalin NH. A dynamic relation between whole-brain white matter microstructural integrity and anxiety symptoms in preadolescent females with pathological anxiety. Transl Psychiat. 2022;12(1):57.
5. Cerghet M, Skoff RP, Swamydas M, Bessert D. Sexual dimorphism in the white matter of rodents. Journal of the Neurological Sciences. 2009 Nov 15;286(1–2):76–80.
6. Yasuda K, Maki T, Kinoshita H, Kaji S, Toyokawa M, Nishigori R, et al. Sex-specific differences in transcriptomic profiles and cellular characteristics of oligodendrocyte precursor cells. Stem Cell Res. 2020;46:101866.
7. Swamydas M, Bessert D, Skoff R. Sexual dimorphism of oligodendrocytes is mediated by differential regulation of signaling pathways. Bansal R, Macklin Wendy B, Vellis J de, editors. Journal of Neuroscience Research. 2009 Nov 15;87(15):3306–19.